Phase I, Open Label Safety Study of Ublituximab for the Treatment of Acute Neuromyelitis Optica Relapses

Michael Levy, MD, PhD and Maureen A. Mealy, RN, BSN

[1]Department of Neurology, Johns Hopkins University, Baltimore, MD, USA

Background

The current standard of care for treatment of acute NMO attacks of both optic neuritis and transverse myelitis is a 5-day course of high dose methylprednisolone (1000 mg daily). In some patients, this course of steroid treatment is sufficient to suppress CNS inflammation and reverse some neurologic dysfunction but in many patients, steroids alone are not sufficient to suppress CNS inflammation, and treatment escalation to plasma exchange is necessary within 5-7 days.

The rationale for using ublituximab, a novel glycoenghineered anti-CD20 monoclonal antibody, in this patient population is based on the known roles of B cells, antibody production and plasma cells in the pathophysiology of NMO. NMO is characterized by the presence of an anti-AQP4 antibody, which are produced by differentiation of B cells to plasmablasts. B cells also play a role as potent antigen presenting cells. The strongest evidence of the importance of B cells in NMO comes from studies of B cell depletion, most commonly with another anti-CD20 monoclonal antibody, rituximab (Rituxan®). Rituximab has been shown in multiple retrospective and prospective studies to be effective in reducing NMO relapse risk up to 83% and achieving remission solely by depletion of circulating CD20+ B cells, despite no change in plasma cell population and anti-AQP4 antibody titers. These human studies strongly suggest a critical role for B cells in the pathophysiology of human disease.

While typically used in the prevention of disease, B-cell depletion may be beneficial in the treatment of an acute relapse as well. Emerging evidence indicates that peripheral B cells are activated during a relapse and plasmablast production of anti-AQP4 antibodies spikes. B cells are also found within acute lesions of the spinal cord and optic nerve suggesting roles both in the blood and in the central nervous system during a relapse.

Objective

To assess safety of acute B cell depletion in NMO subjects with acute relapse of optic neuritis or transverse myelitis who are treated with ublituximab + steroids versus steroids alone beginning on dose administration and ending with recovery of B cells.

Methods

This is a Phase 1 open-label, add-on, single treatment arm, unblinded, single center interventional trial in NMO/NMOSD patients in which experimental subjects will receive one (1) infusion of 450 mg of intravenous ublituximab at the onset of an NMO exacerbation in addition to standard of care treatment with daily intravenous methylprednisolone at 1000 mg for five days.

Patients must meet Wingerchuk 2015 criteria, and relapses must present with new symptoms and exam changes attributable to an enhancing MRI lesion. Circulating B cells must number at least 0.5% of total lymphocytes.

The overall objective is to assess the safety of ublituximab as add-on therapy to steroids for treatment of acute optic neuritis and/or transverse myelitis in NMO and NMOSD. Clinical outcomes will be monitored for efficacy using three functional neurologic tests, the Expanded Disability Status Score (EDSS), Timed 25-Foot Walk and High Contrast Visual Acuity (HCVA). These will be assessed days 1 and 5 of hospitalization, on Days 13 and 20 (if necessary) and at follow up in 90 days. B cells are monitored monthly until they replete to at least 0.5%.

Demographics of Subjects

Subject	Age	Sex	Race	NMO diagnosis	Duration of Disease (yrs.)	# of previous relapses	Clinical presentation	Background immunotherapy
1	41	F	Hispanic	Seropositive NMO	13.6	4	Thoracic myelitis affecting lower limb strength	none
2	54	F	African American	Seropositive NMO	6.4	6	Thoracic myelitis affecting sensory function in lower limbs	Mycophenolate (d/c'd 3 mo. prior)
3	48	F	African American	Seropositive NMO	13.3	5	Thoracic myelitis affecting lower limb strengthUnilateral	None (last RTX 3 yr. prior)
4	26	F	African American	Seropositive NMO	8.3	4	Optic neuritis causing vision loss	Mycophenolate
5							To be recruited	

Efficacy: Individual EDSS Scores

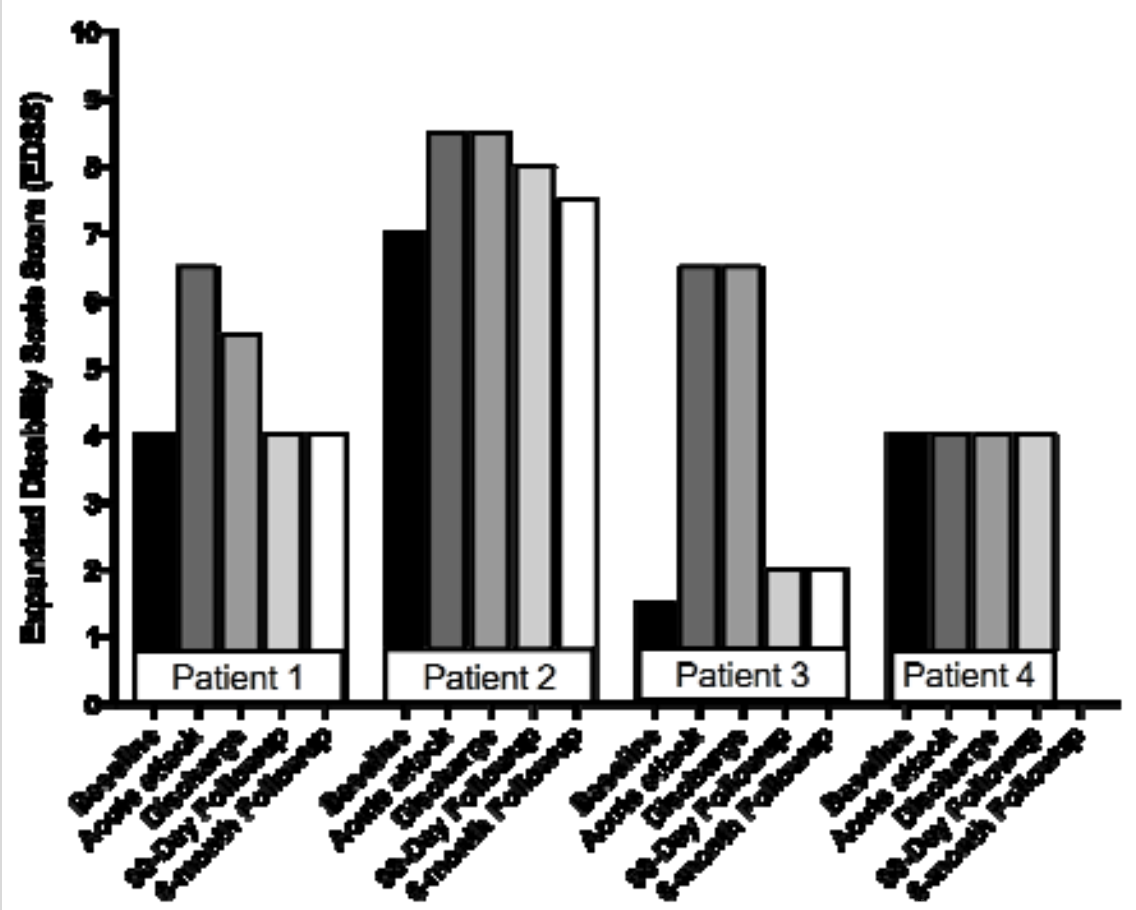
Expanded Disability Scale Score (EDSS)
Patient 1
Patient 2
Patient 3
Patient 4
Baseline
Acute attack
Discharge
90-Day Followup
6-month Followup

Expanded Disability Scale Scores were recorded for each patient at baseline in clinic, at peak of relapse in the hospital, on discharge, and at 90-day and 6-months follow up in clinic. In 3 patients, the EDSS increased from baseline to the acute relapse, and then declined back to baseline or better by 90-day follow-up. Patient 4 suffered optic neuritis from a poor baseline that was not captured by the EDSS rating scale. Patients 2 and 4 required plasmapheresis. Patient 4 has not yet been assessed at 6-month follow-up.

Safety and Secondary Outcomes

Subject	Adverse Events
1	Total body pain for 30 minutes at onset of infusion
2	None
3	Total body pain for 30 minutes at onset of infusion
4	Total body pain for 15 minutes at onset of infusion
5	To be recruited

Timed 25-Foot Walk and B cell counts

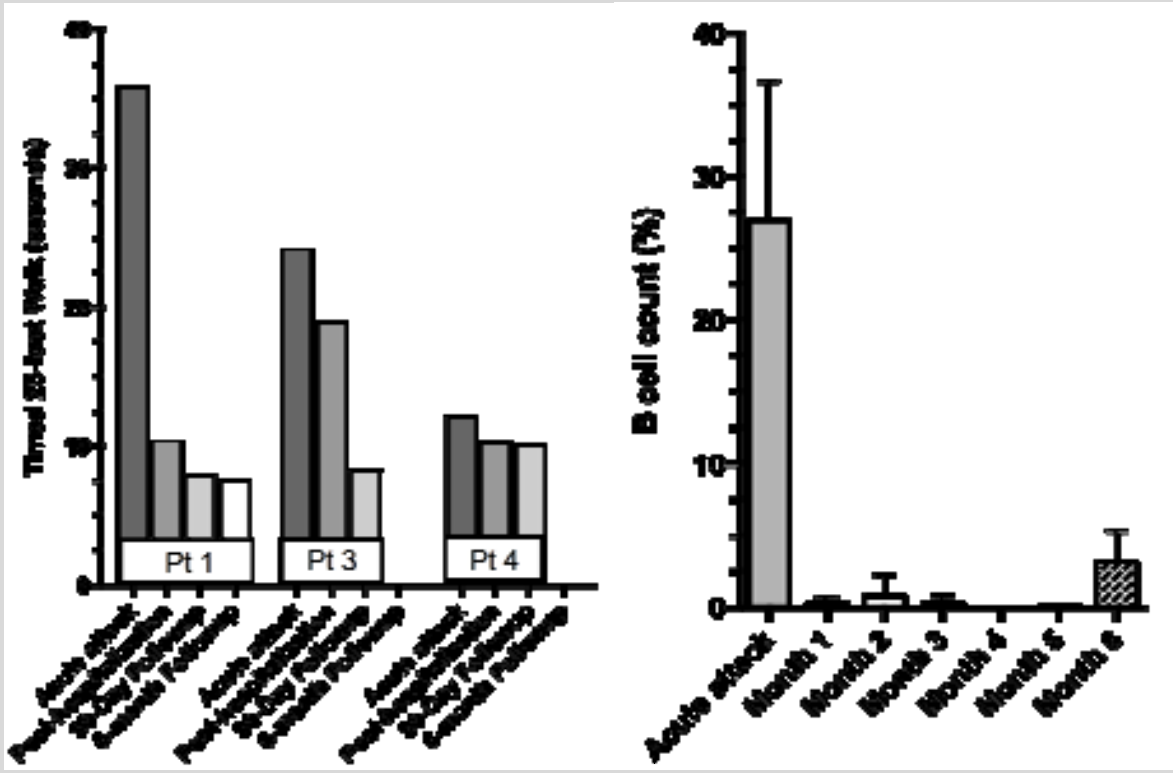
Pt 1
Pt 3
Pt 4
B cell count (%)
Acute attack
Month 1
Month 2
Month 3
Month 4
Month 5
Month 6

Conclusions

- **Ublituximab effectively depletes B cells during an acute NMO relapse without significant risks in this patient population**.
- Disability from NMO relapses increases acutely from baseline at the time of presentation, and drop over time after treatment with ublituximab along with standard of care corticosteroids. A phase II/III study with a placebo control is necessary to confirm efficacy.
- Two of four subjects repleted their B cell counts earlier than the expected 6 months time period. This is perhaps related to the lower dose and frequency of ublituximab compared to the reported experience with rituximab.
- Timed 25-foot walk and visual acuity, as NMO-specific functional measures of recovery, improved along with EDSS scores in this study.

Funding

- This work was funded by an investigator-initiated trial grant from TG Therapeutics, the manufacturer/distributor of ublituximab.